Does Upregulation of Limb Perfusion and Effective Tissue Micro Deformation Increase Wound Healing During Textile Compression Therapy

Martin J Winkler Sr, MD, FACS • Creighton University, Omaha Nebraska
Linda T Miller, PT, DPT, CLT, CWT • Keystone Care Homecare and Hospice, Wyndmoor, Pennsylvania

Non Healing Arterial Ulcer in Profoundly Ischemic Leg with NO Doppler Signals Below Knee

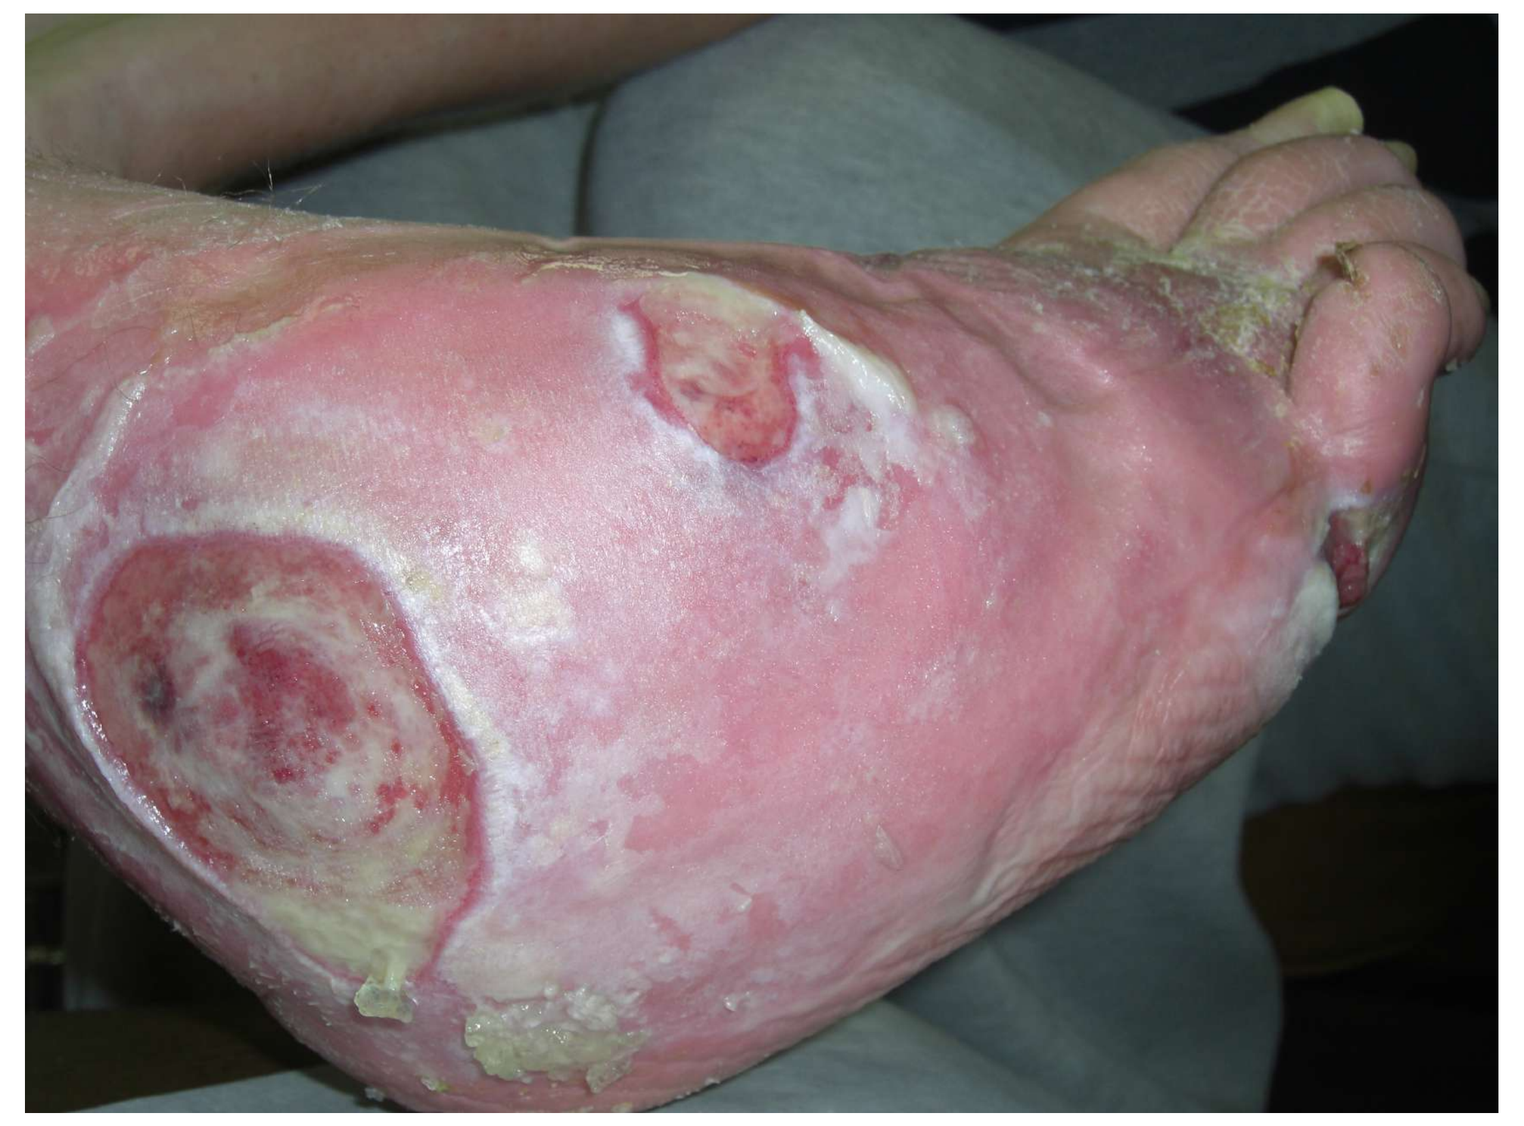

Rx Day #0:
54 yo female with L BKA, Hx 2+ packs/day smoker, aortic occlusion below renal arteries with 4 painful ischemic ulcers and right lateral malleolar bone exposed. Extensive skin maceration is the result of polyethylene sheet over lateral malleolus during NPWT. No doppler signals below knee; patient refused amputation.

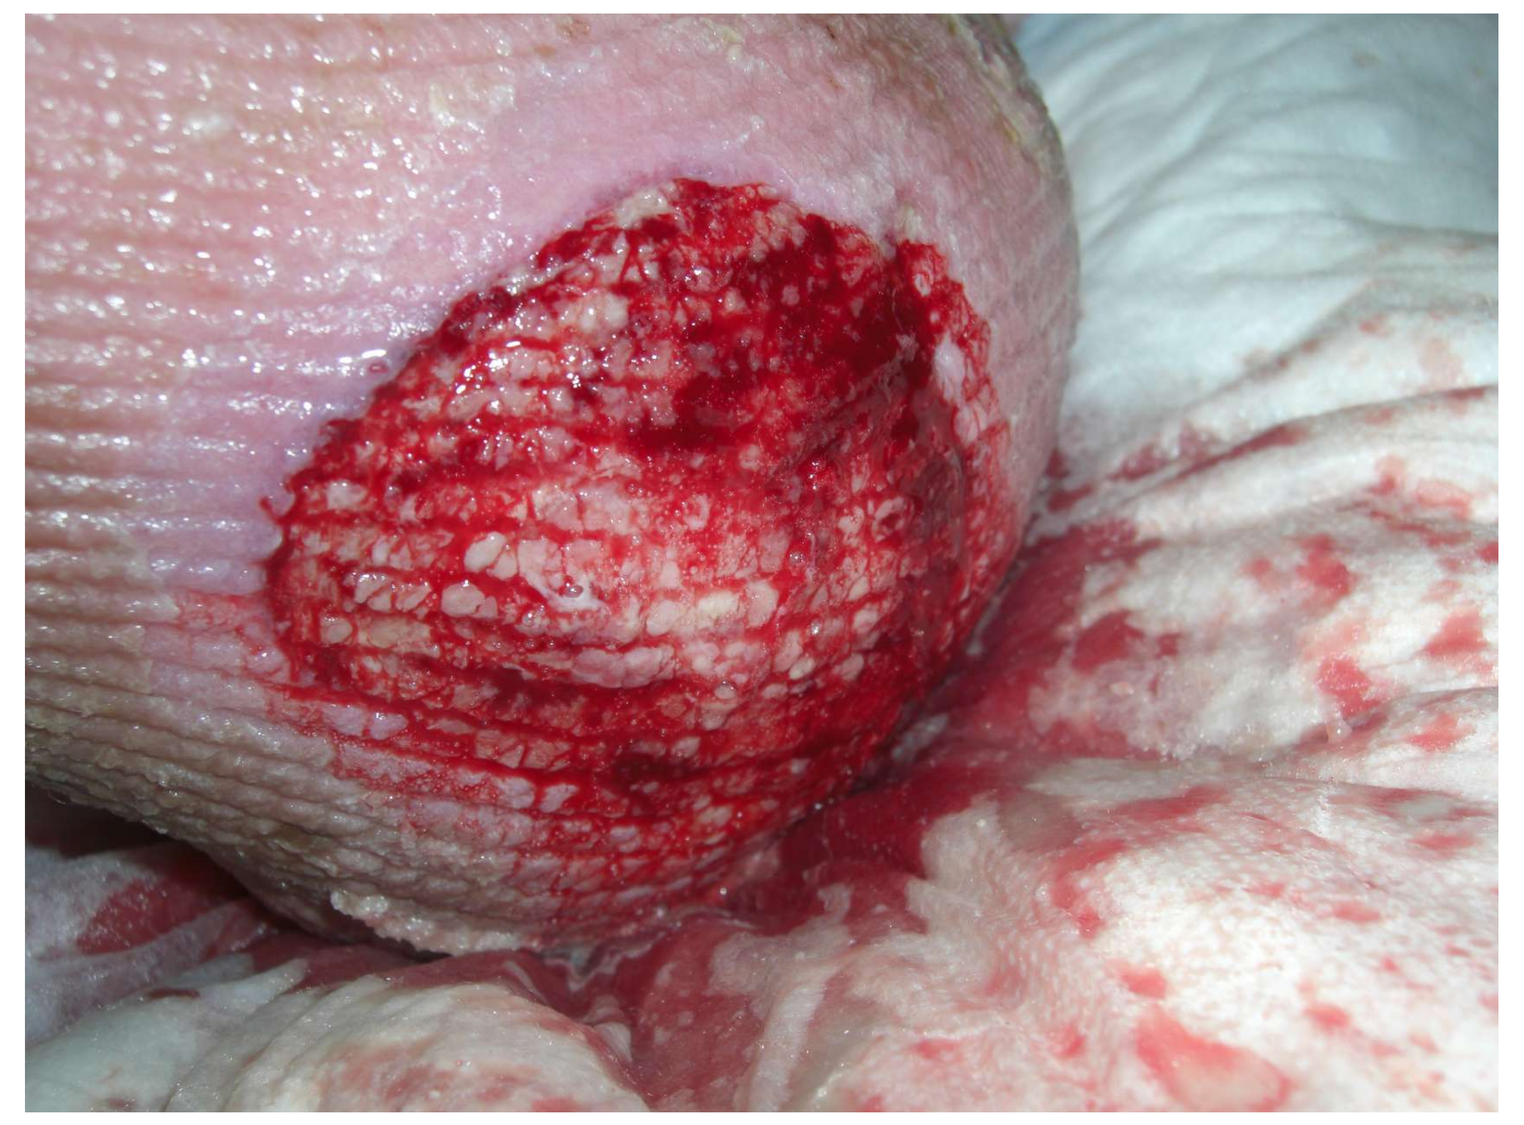

Rx Day ~ #45:
Dramatic wound bed progress, robust neo epithelization, healthy new normal appearing skin at wound margins and bleeding, which all suggest improved perfusion. Fuzzy wales deliver static Newtonian force creating furrows in the skin, we posit turbulent flow in 7μm arterials beneath furrows stimulates release of nitric oxide (NO) that vasodilates downstream arterials.

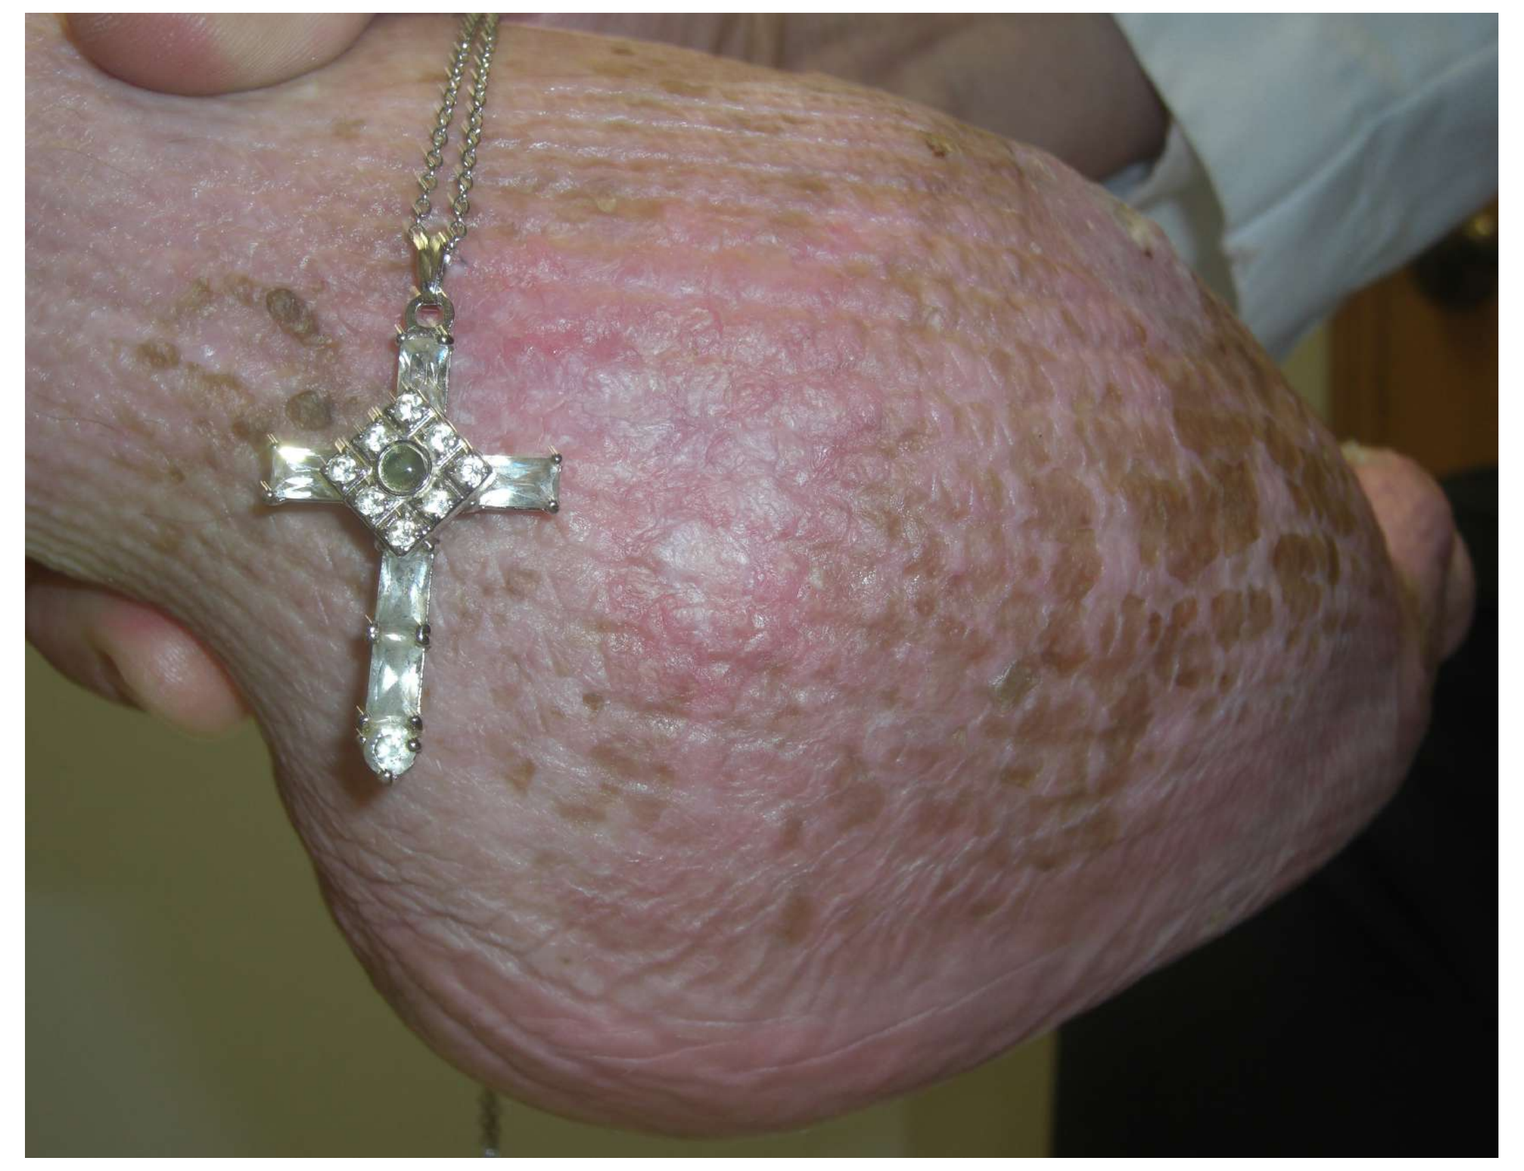

Day #193:
Pt discharged. Observe ischemic full thickness wound healing without scar—a bedside clinical sign of regenerative stem cell healing and is commonly seen with Negative Pressure Wound Therapy (NPWT). Ehmann (2022) published the observation that robust regenerative healing seen with fuzzy wale compression* is almost certainly the result of Positive Pressure Wound Therapy (PPWT) comprising three unique physiologic therapies: 1) enhanced skin perfusion - (probable upregulation of Nitric Oxide), 2) rapid edema control, and 3) tissues experiencing micro deformation in furrows beneath fuzzy wales send mechano transduction signals to their DNA to upregulate gene expression and synthesize the myriad proteins required to heal the wound.

Bilateral Refractory Mixed Venous Leg Ulcers

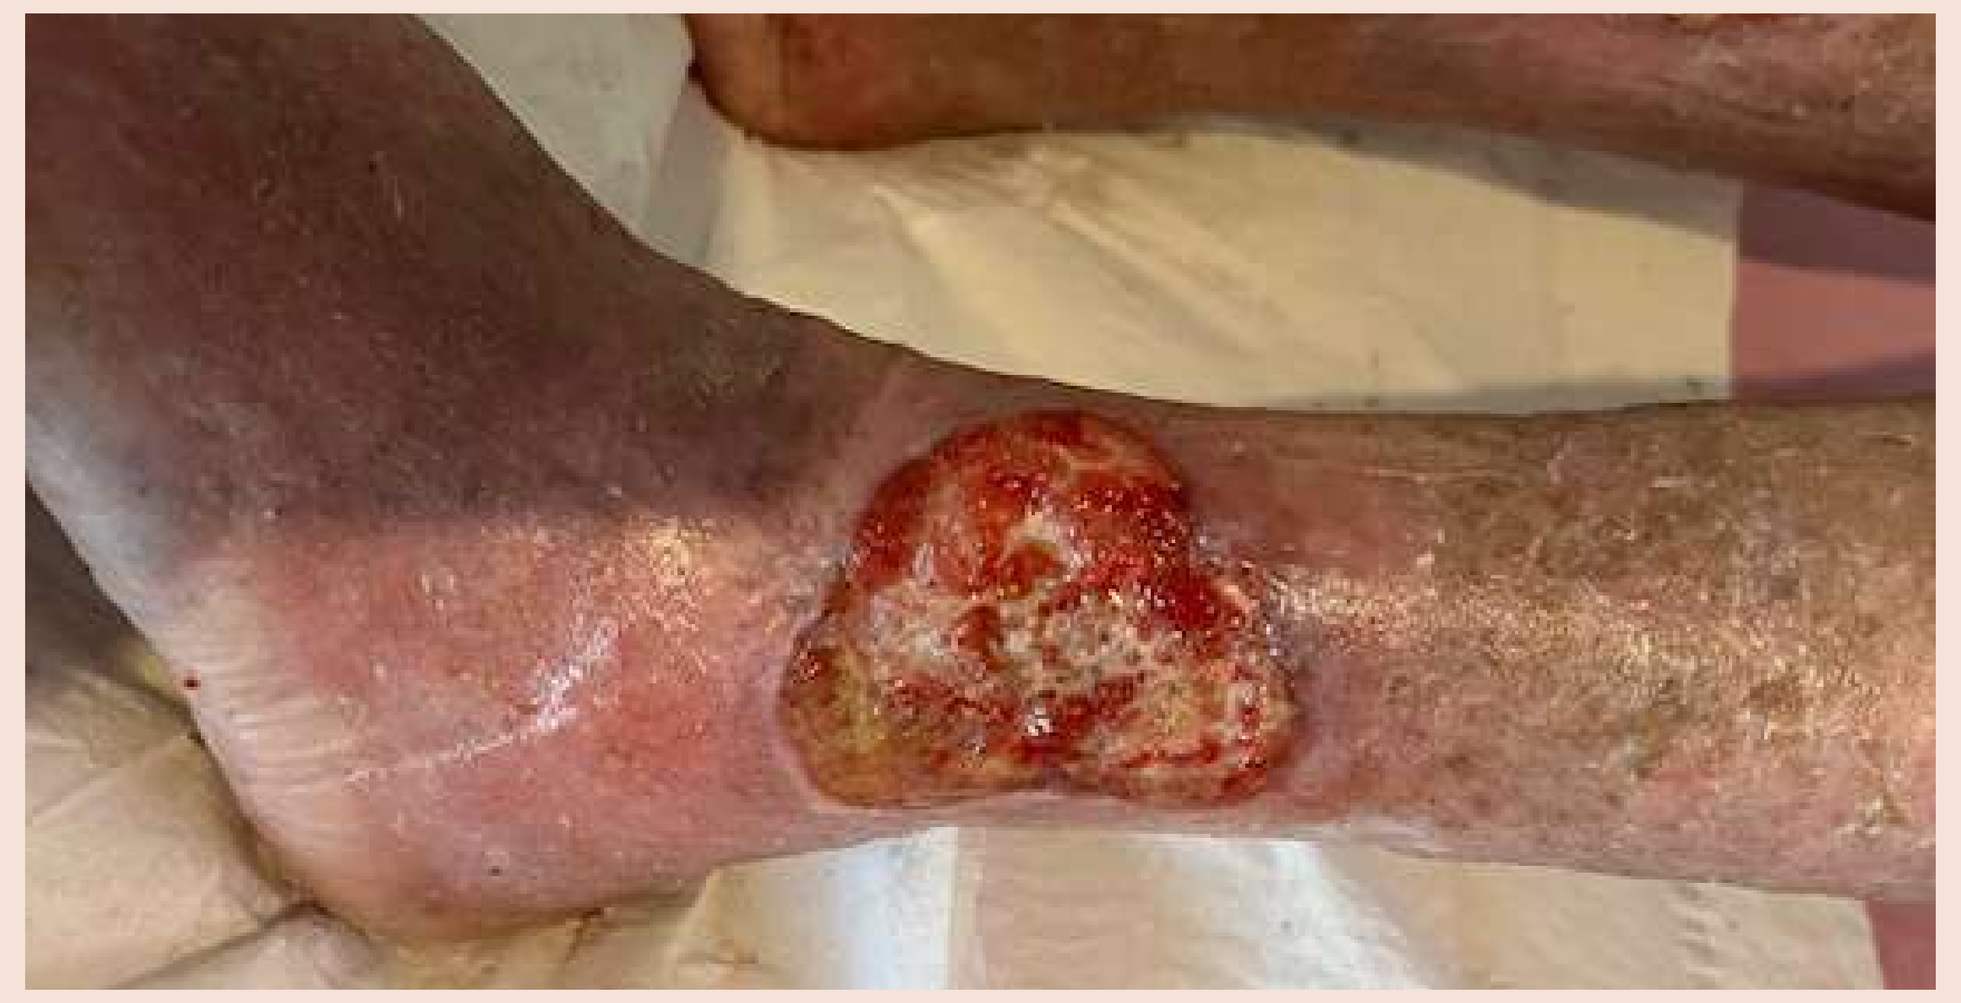

Rx Day #0:
Refractory painful Left Leg Mixed Venous Ulcer x 3 1/2 years. Observe painful peri wound dermatitis, wound edge epiboly (swelling), and exuberant, ragged, friable wound granulation tissue. (Photo © Linda Miller, Wyndmoor, PA)

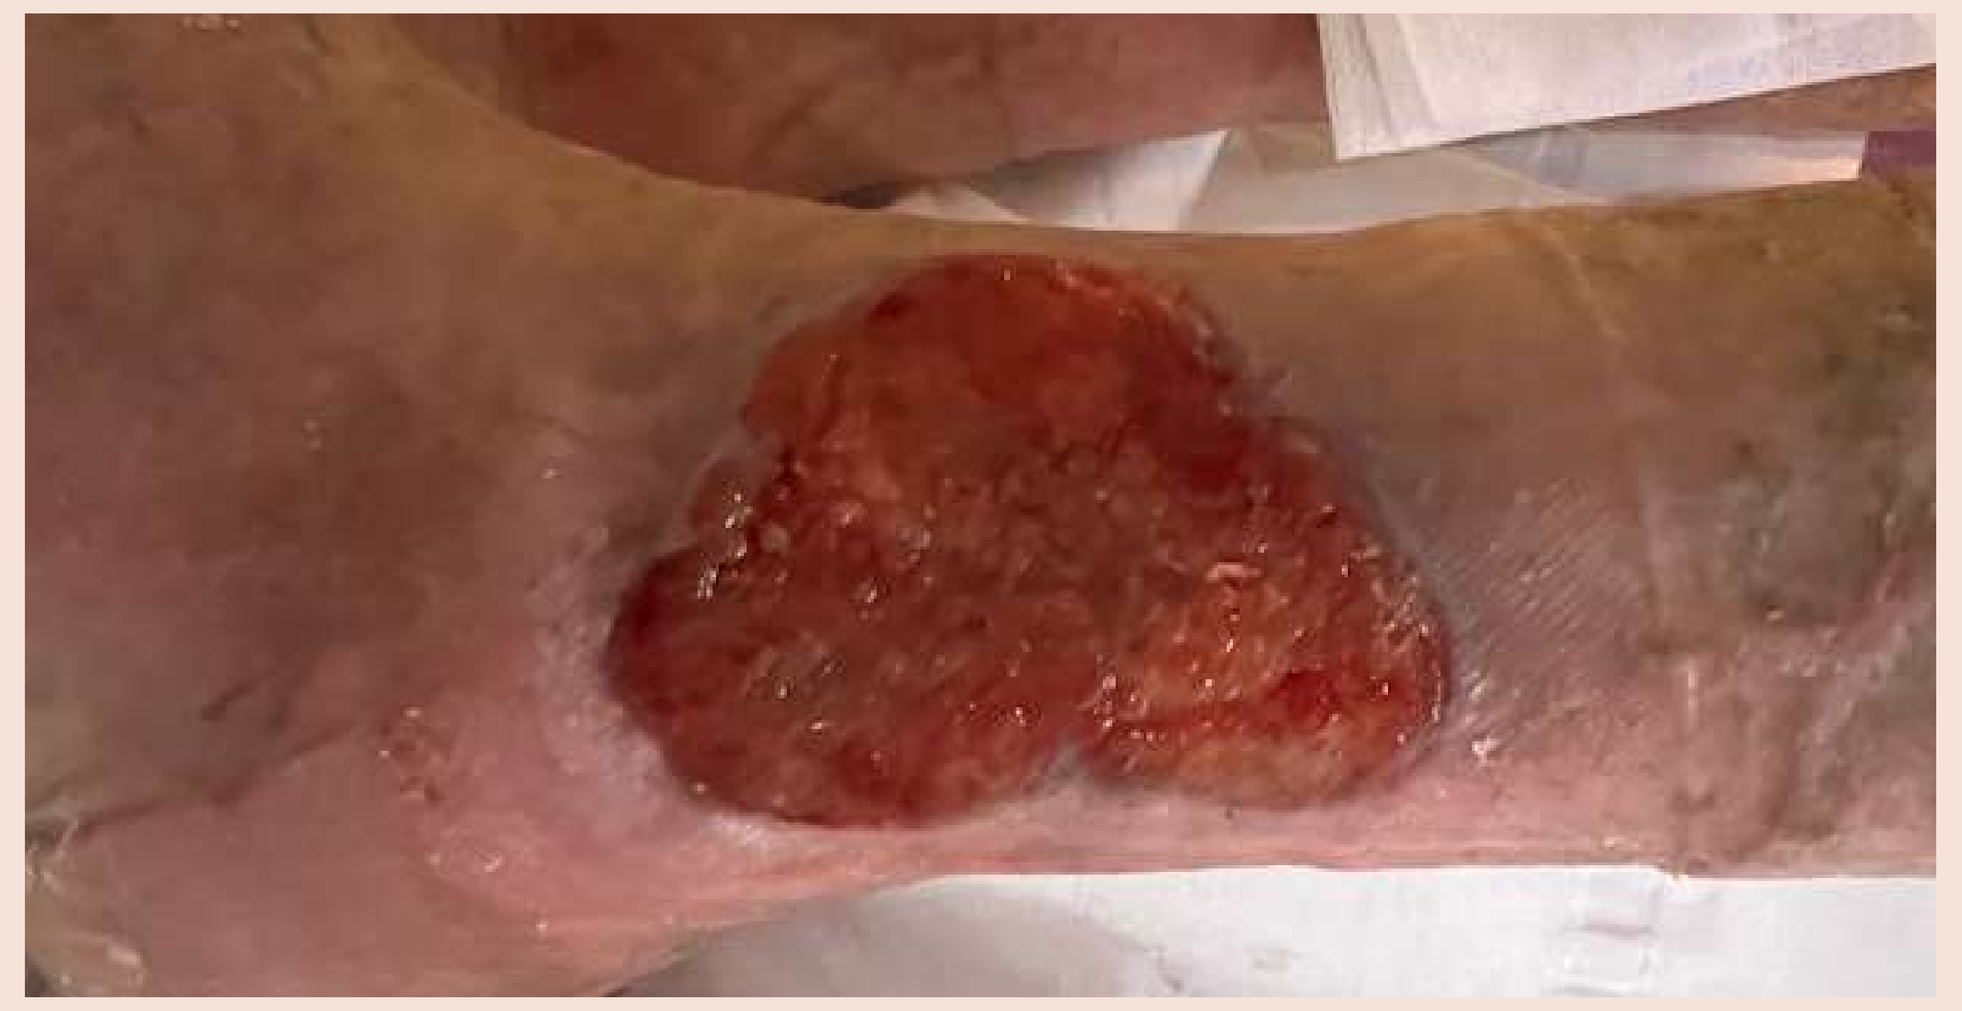

Rx Day #7:
of highly absorptive cloth and two layer compression. Note periwound stasis dermatitis and robust granulation tissue. (Photo © Linda Miller)

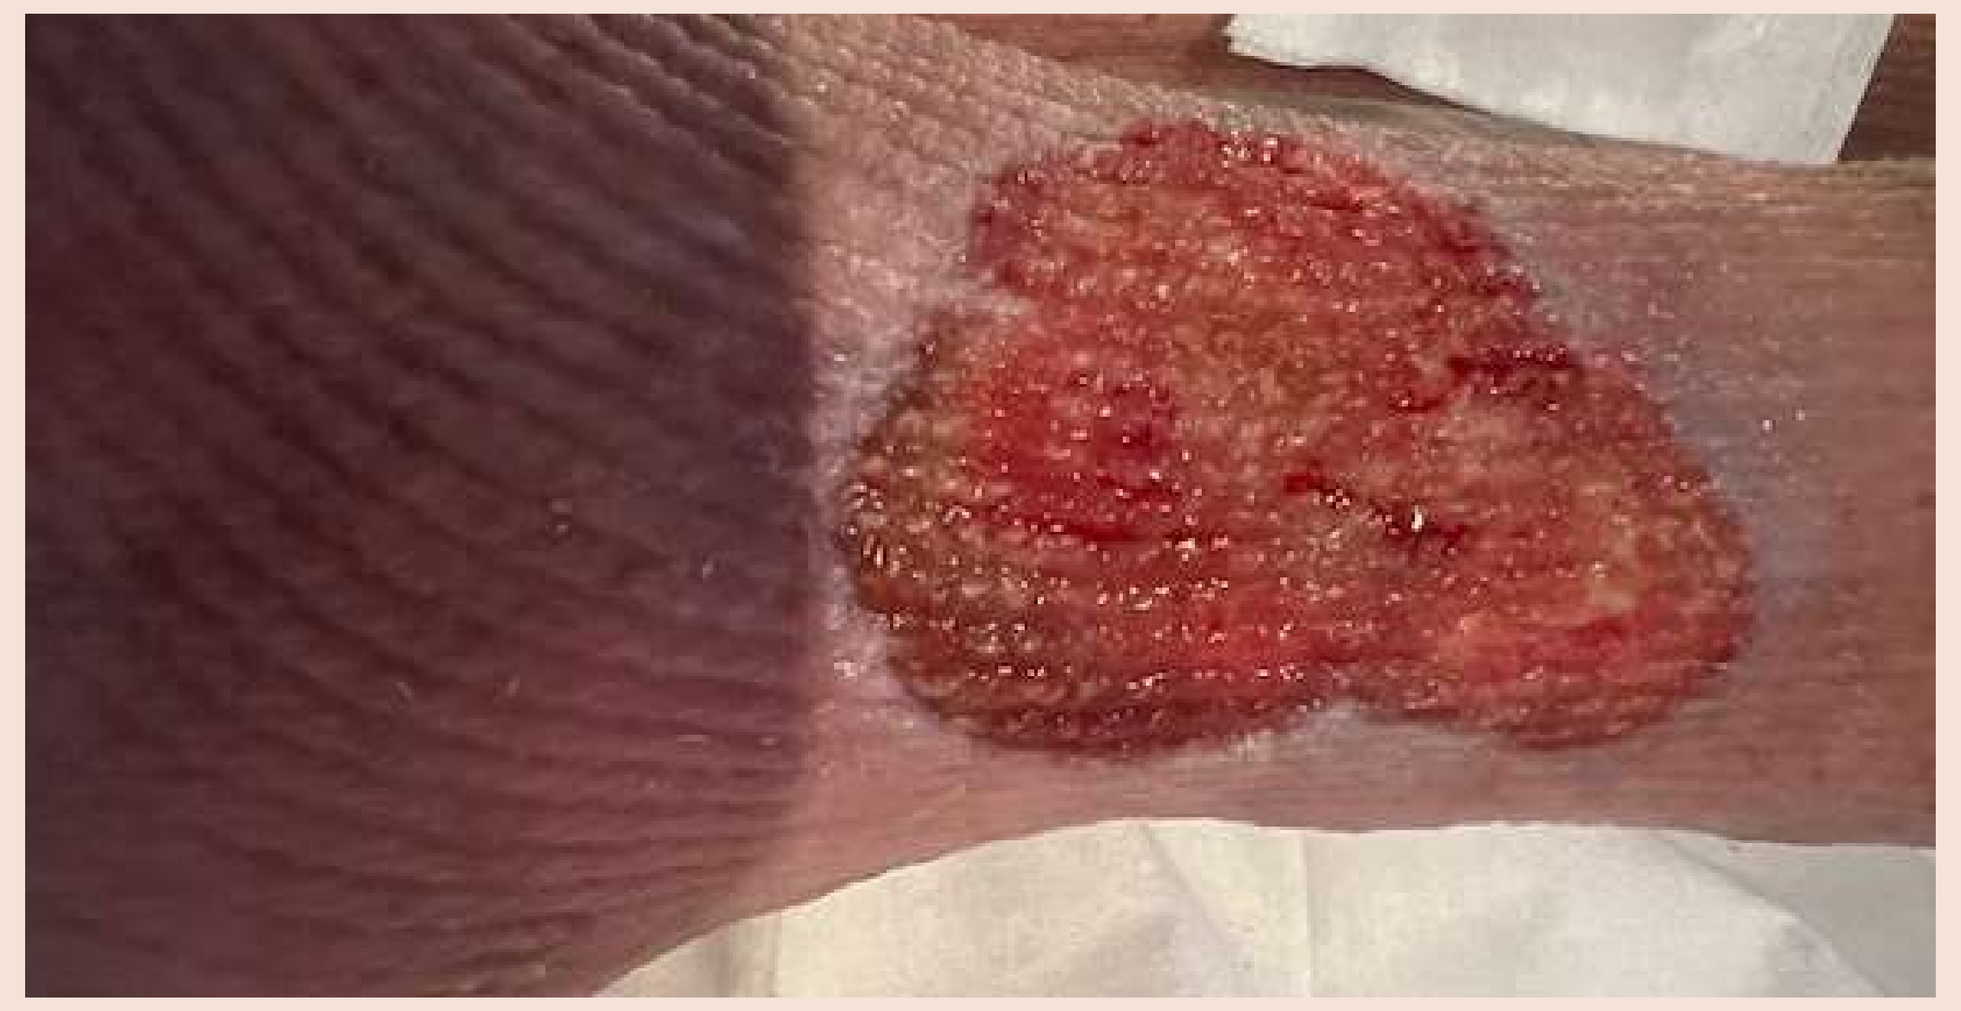

Rx Day #14:
of Fuzzy Wale Elastic Compression Stockinet* as the wound contact layer. Observe dramatic improvement of periwound dermatitis, decreased wound edge epiboly, less friable granulation, and less pain. (Photo © Linda Miller)

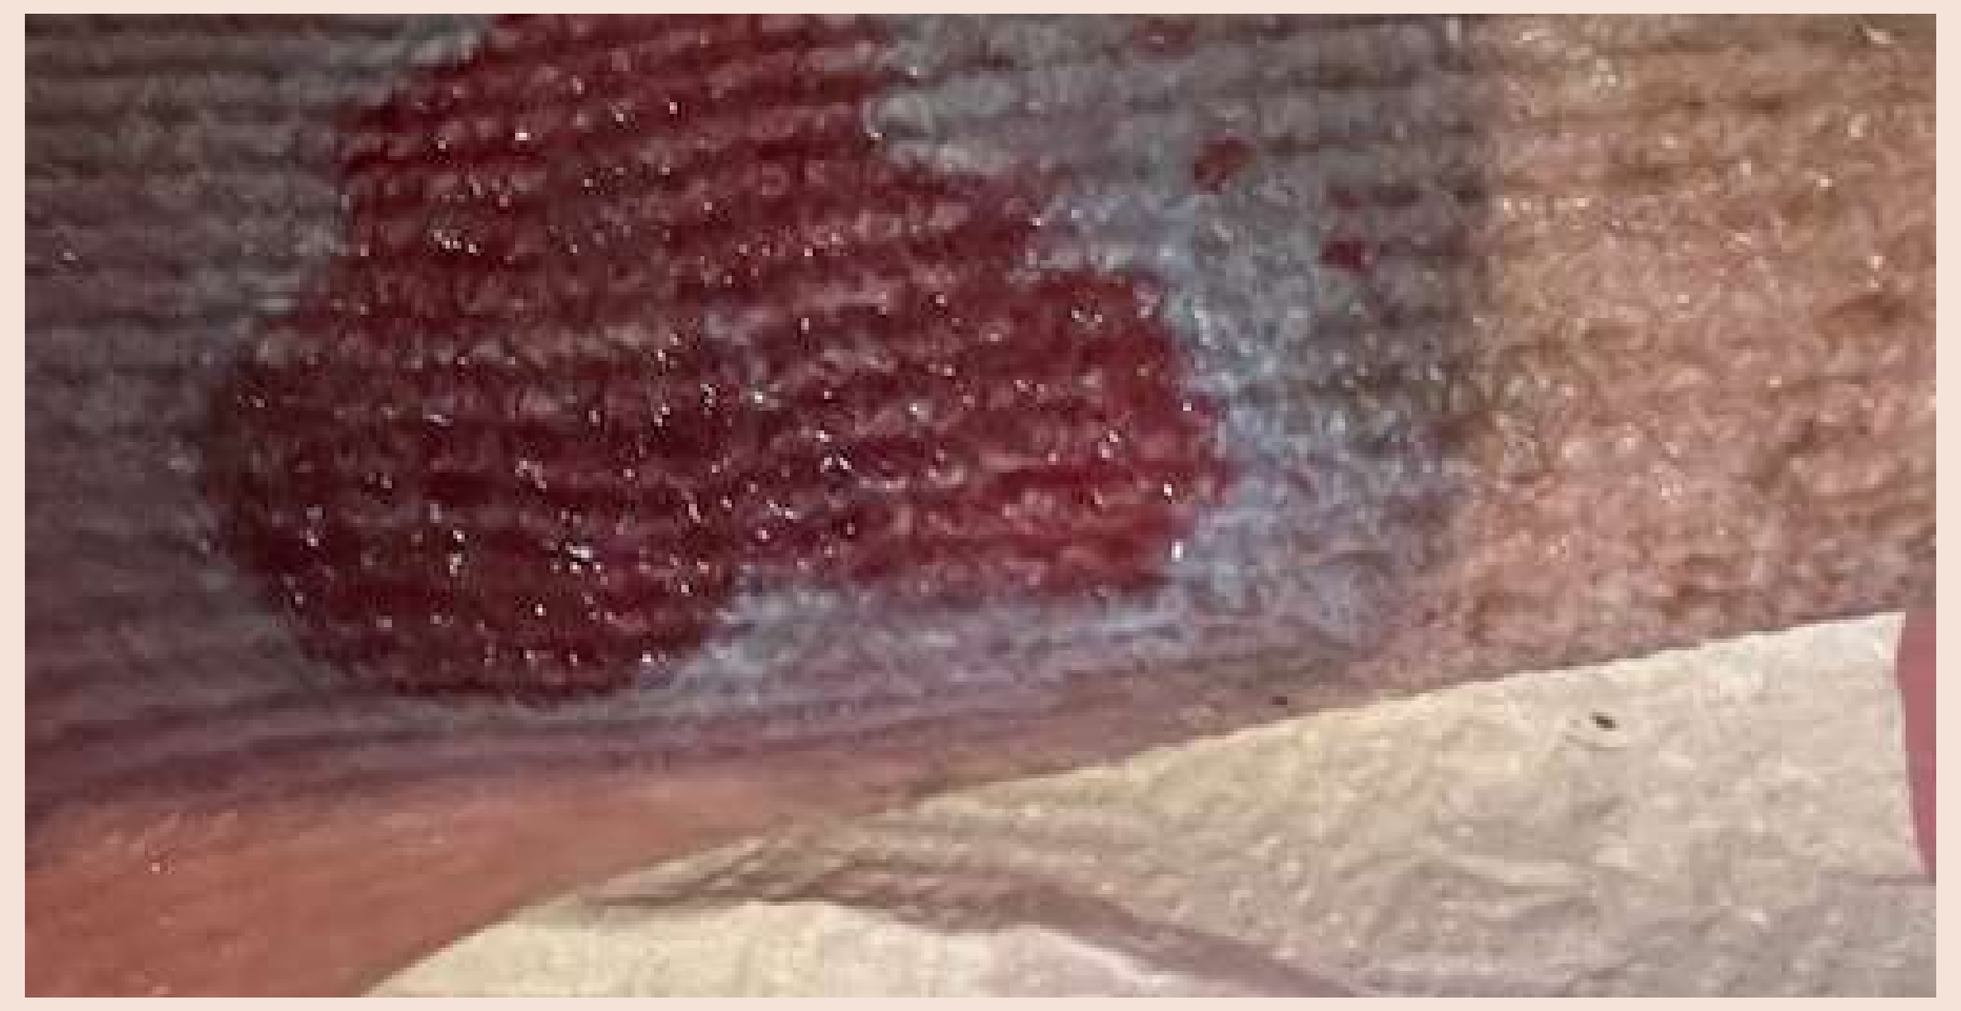

Rx Day #35:
of Fuzzy Wale Elastic Compression Stockinet. What looks like a dusting of snow on a red earth cornfield, are epithelial cells moving across the wound on granulation tissue between furrows. This patient will go on to heal a full thickness chronic wound without skin grafting, which in itself is a miraculous thing. (Photo © Linda Miller)

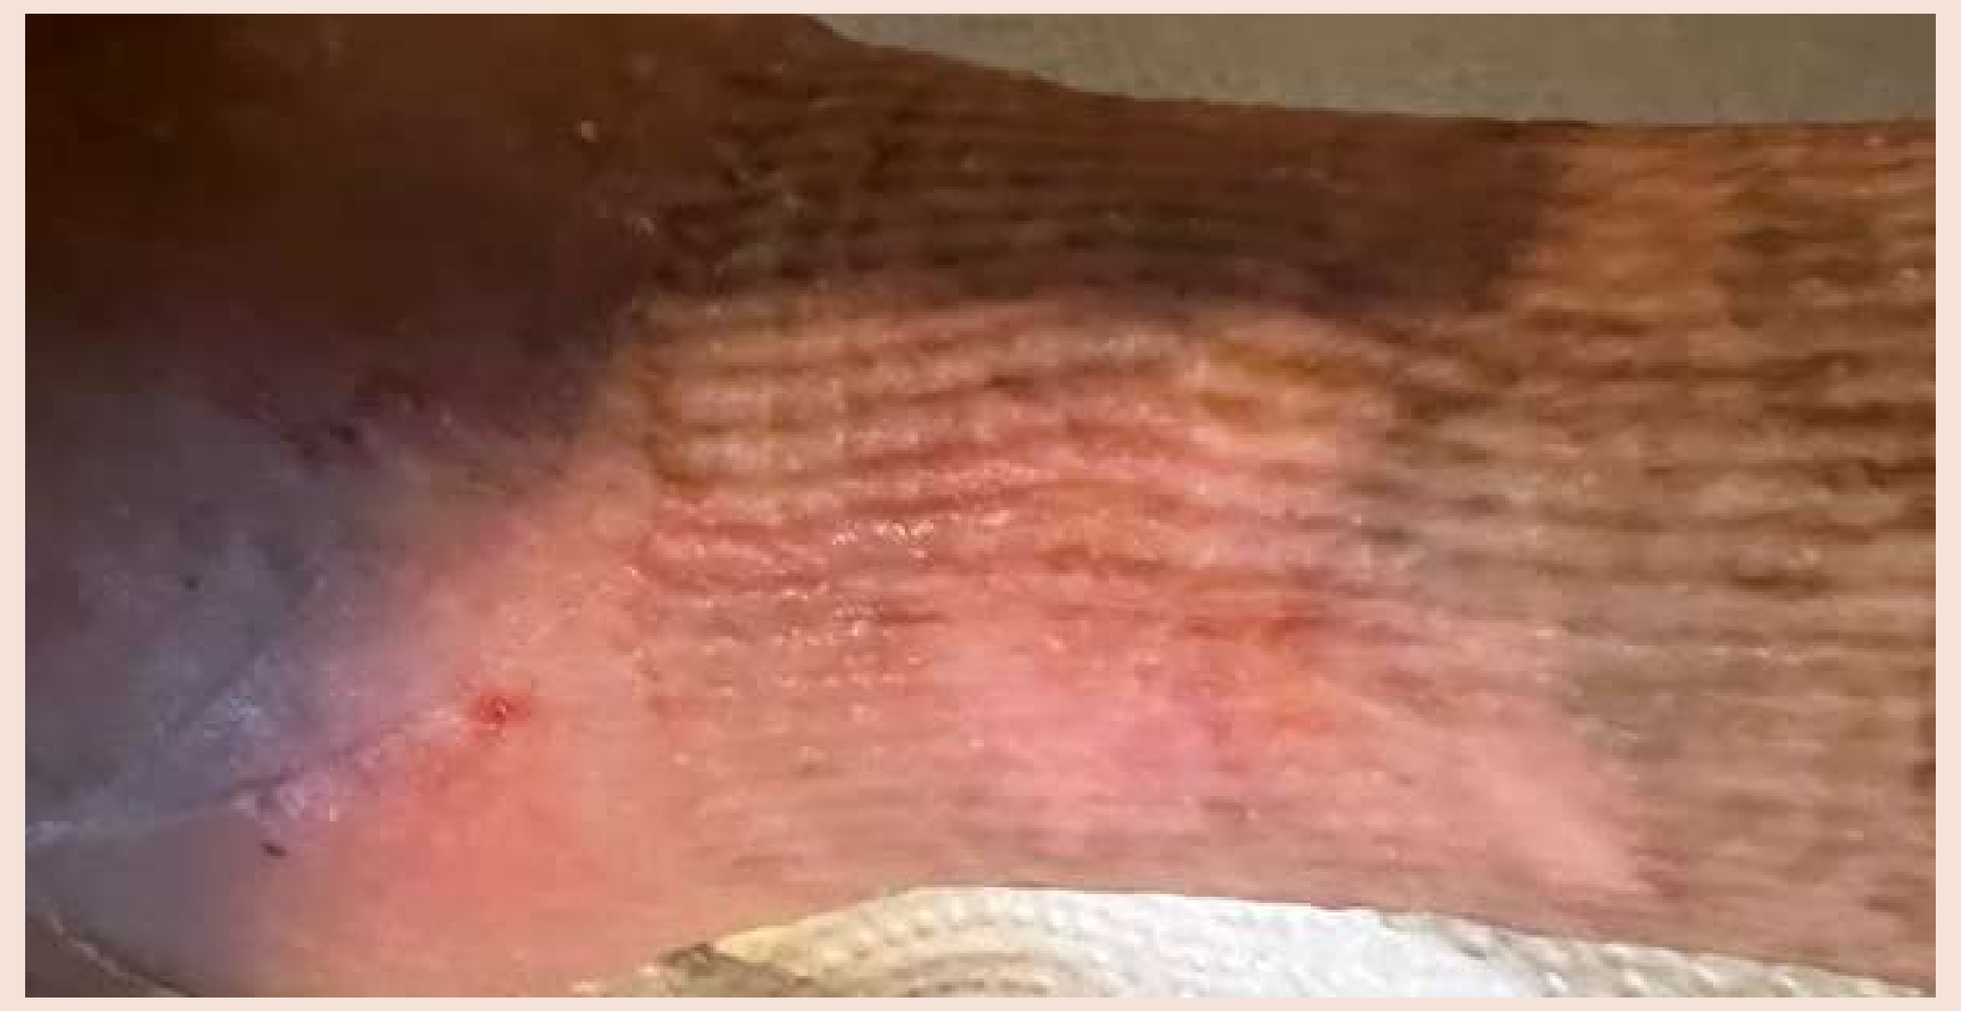

Rx Day #49:
of Fuzzy Wale Elastic Compression Stockinet. Photo of the healed wound reveals healthy new skin with no evidence of the prior wound. Importantly, healing without scar is a bedside sign of regenerative healing, which is also seen with 3M's open cell foam delivering vacuum pressure to wound surface. (Photo © Linda Miller)
(nota bene treatment day numbers are approximations)

* EdemaWear® Elastic Compression Stockinet, Compression Dynamics LLC, Omaha, Nebraska 68102

Painful, Refractory, 4 Years of Wound Center Care, Mixed Etiology Ankle Ulcer

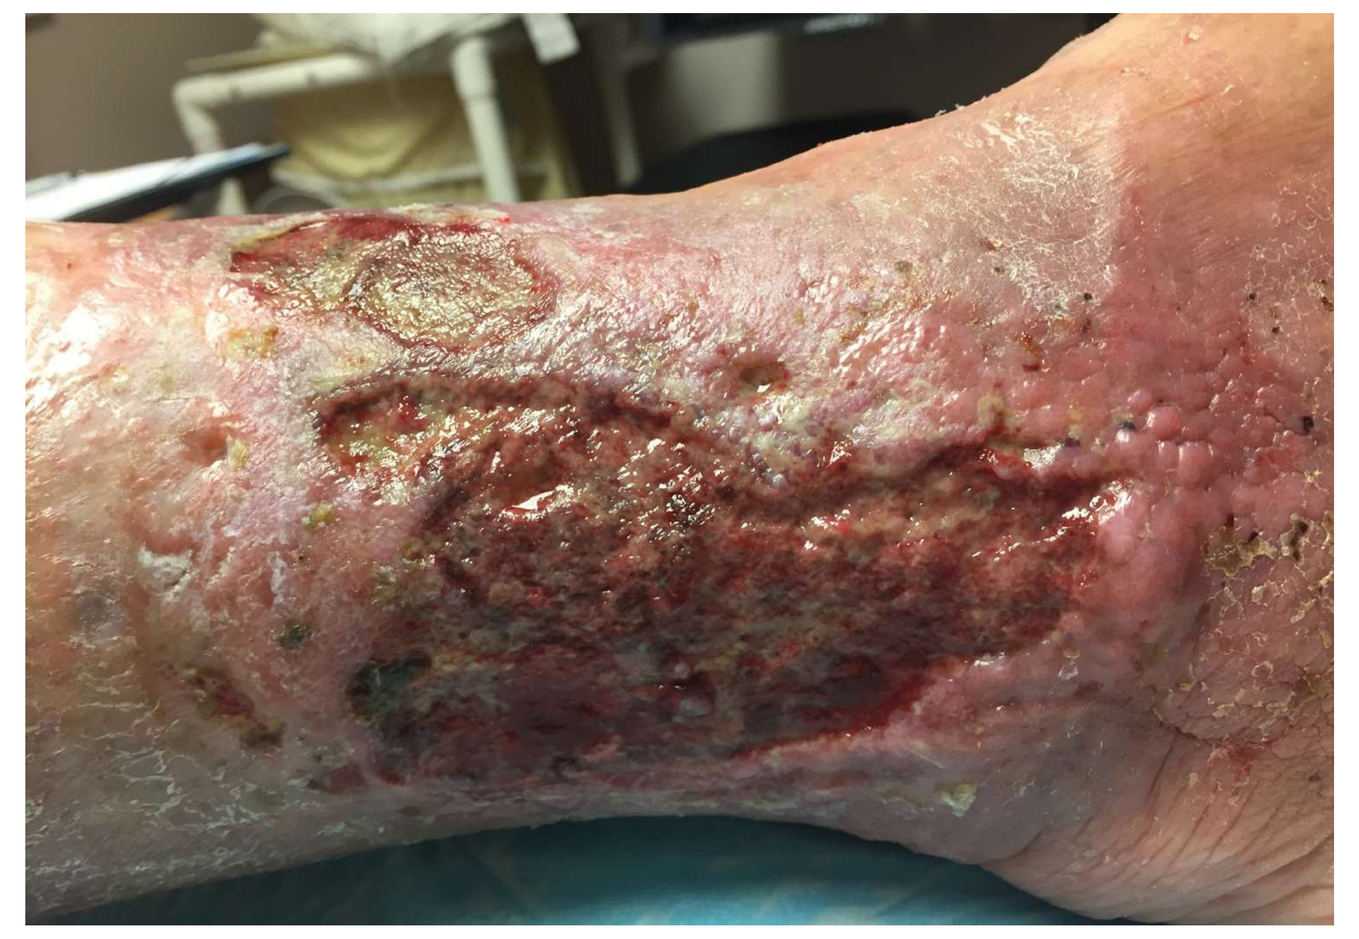

Rx day #0:
Observe extensive nodular periwound dermatitis with areas of painful nodular edema, 3-5 mm wound edge epiboly, and shaggy wound bed granulation tissue weeping exudate/transudate.
(Photo © Marta Ostler, Sheridan, WY)

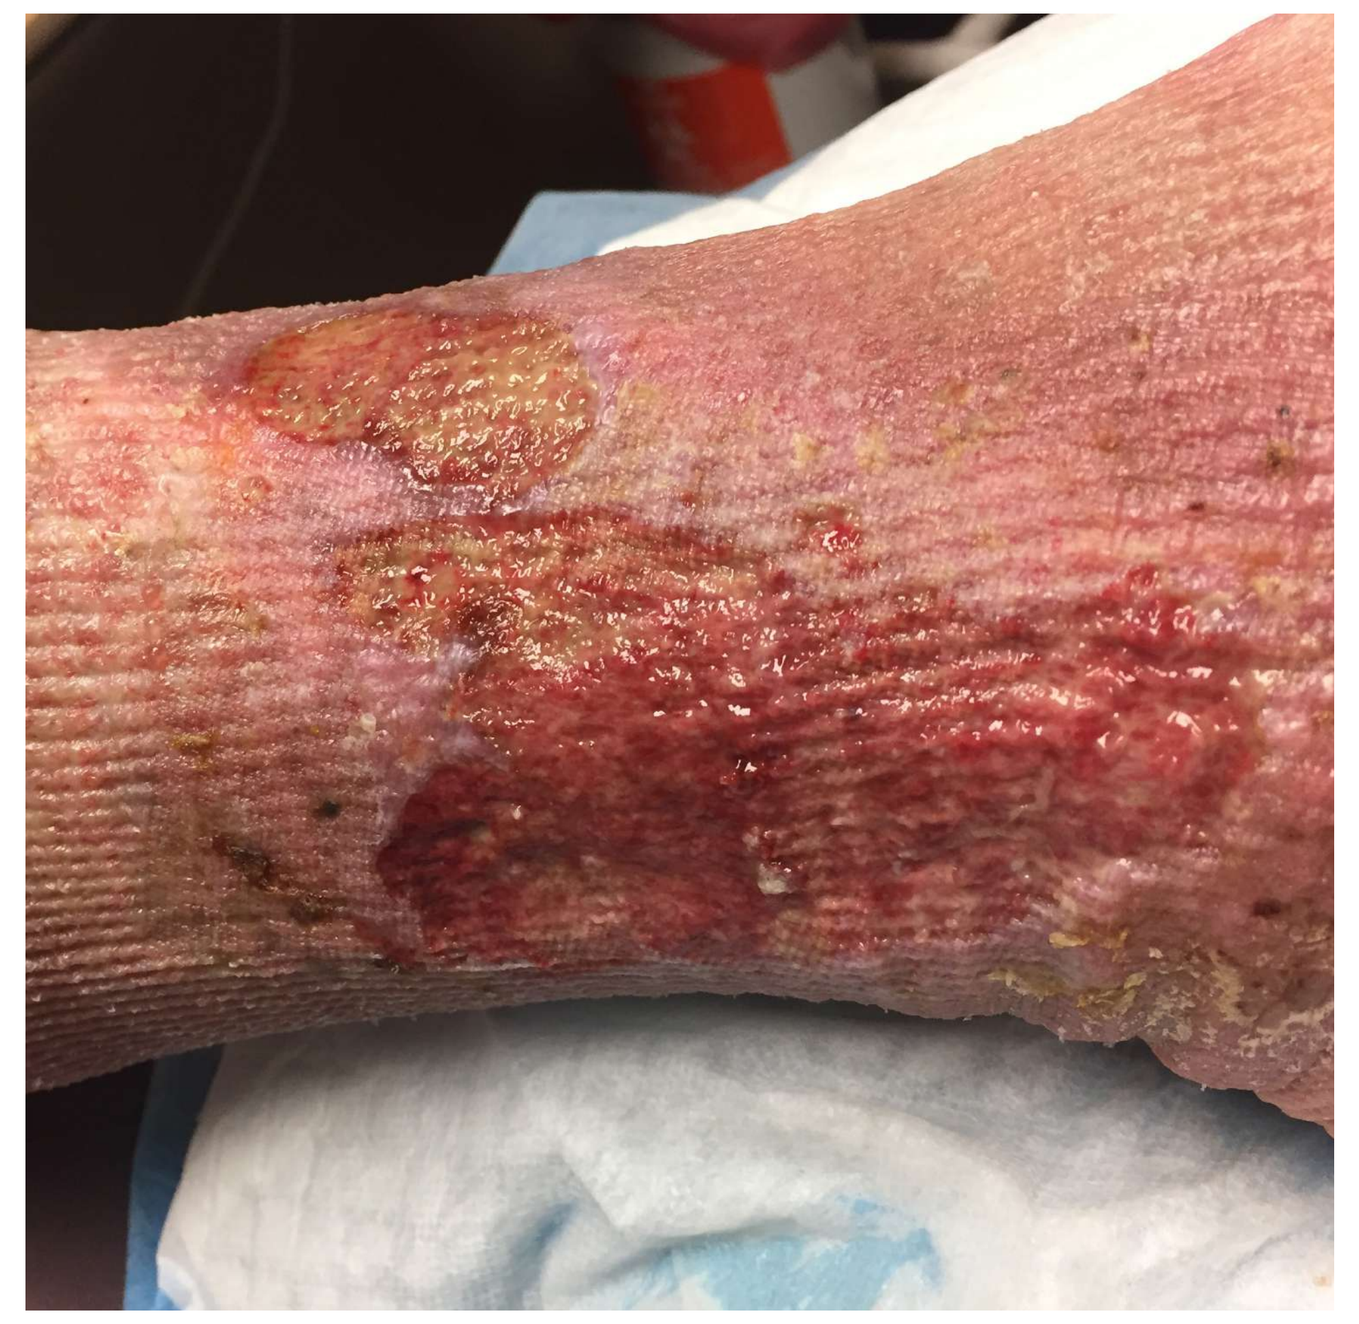

RX Day #9:
Clinic Visit #3: Observe corn row furrows have formed beneath fuzzy wales in the skin and wound granulation tissue, dramatic resolution of painful nodular periwound stasis dermatitis, resolution of wound edge epiboly, and dramatic wound bed progress. What is not seen are the smiles on the patient and his family.
(Photo © Marta Ostler)

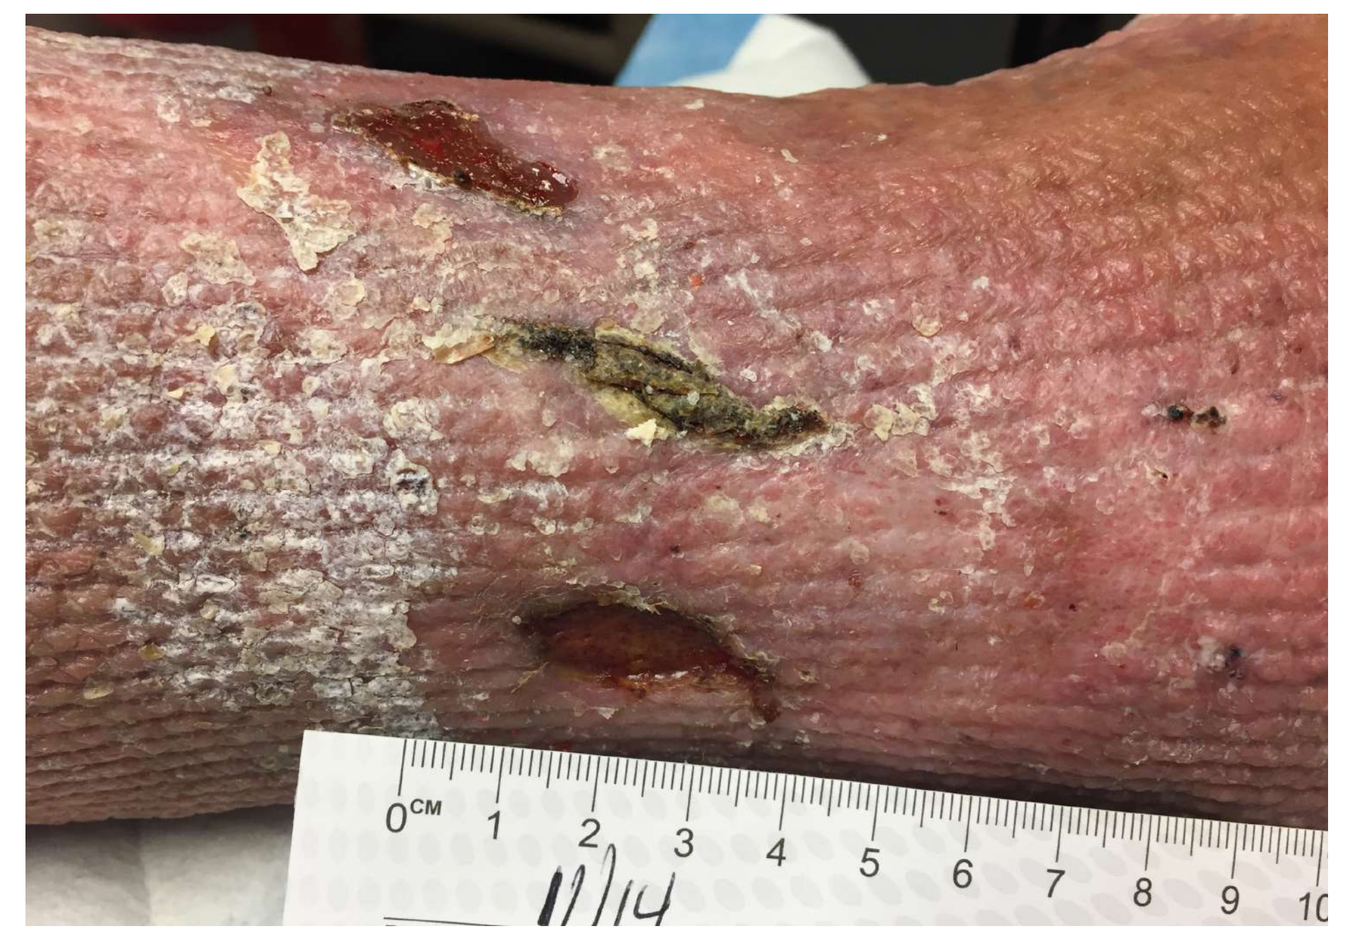

Day #106:
Patient discharged to primary doctor. Observe wound appears 95% healed, it is difficult to see the outline of the original wound edge, healing without scar is a sign of regenerative healing. Fuzzy wales deliver static force which creates cornrow furrows in skin and winkled subcutaneous fat as a result of stretched spandex elastomer. Regenerative healing is seen frequently when fuzzy wale compression stockinet is used in intimate contact with the skin and wound bed in a manner similar to regenerative healing seen in Negative Pressure Wound Therapy (NPWT).
(Photo © Marta Ostler)

Introduction
Textile compression has been used to treat wounds over 4 millennia, with Asklepion priests swaddling leg ulcers 350 BCE. Mechanisms underlying compression's efficacy remain poorly understood. We present 3 cases of refractory non-healing wound outliers treated with a novel knitted compression stockinet. Textile fuzzy wales create parallel, longitudinal furrows in skin and subcutaneous fat. Potential mechanisms by which this stockinet may increase perfusion and speed healing are presented.

Methods
Three leg wounds that had previously failed >3 years' wound center treatment (pain preventing prior compression in 2 patients) were treated with Fuzzy Wale elastic Compression stockinet* (FWC), novel in that vertical fuzzy wales compress just 25% of the limb surface, delivering mild 8-12 mmHg compression when measured via standard human compression manikin. (1, 2)

Photographs document clinical progress, including decrease of peri-wound epiboly and painful nodular dermatitis, with robust wound bed progress and rapid neo epithelization. (3, 4)

Results
Case 1: 54 y/o female with vasculopathy, aortic occlusion below renal arteries, s/p left BKA, right pedal doppler signals were absent, with 4 ulcers and exposed lateral mallear bone. FWC rapidly decreased ischemic pain and wound maceration. Photos document scarless regenerative healing at day 193.

Case 2: 61 y/o male with painful mixed bilateral leg ulcers refractory to standard compression, responded dramatically to FWC.

Case 3: 67 y/o male with macerated painful mixed ulcer on right ankle could not tolerate compression over 4 years attending wound clinic. Dramatic wound bed progress documented at day 9 of FWC treatment; discharged Day 106.

Discussion
We propose two potential mechanisms of action for FWC-driven healing. When pedal doppler signals are absent, wound healing is essentially impossible. Textile compression, particularly FWC, may increase perfusion to the compressed area. This may involve stimulating more turbulent flow in small-diameter vessels which increases nitric oxide, release a vasodilator, in cutaneous arterioles. (2, 5)

FWC may contribute to wound healing via tissue micro deformation, tissue experiences simultaneous compression and tension beneath vertical wales. Cell membrane enzymes respond to mechanical forces by signaling their nucleus (6) to increase protein synthesis. (1, 5,7) Further research and textile design is needed.

References
1. Ehmann, S. "Augmented Compression Applications Provide Positive Pressure Wound Therapy (PPWT)" Science Poster, Spring SAWC April 7, 2022, https://edemawear.com/wp-content/uploads/2023/05/spatial-variation-in-tissue-dielectric-constant-tdc-values-along-the-leg-of-healthy-young-adult-females-associated-with-compression-bandaging.pdf
2. Melin, M. "Utilization of Fuzzy Wale Compression Stockinet to Facilitate Healing of Complex Wounds," Fall SAWC, Virtual 2019. https://edemawear.com/wp-content/uploads/2019/04/The-Use-of-Fuzzy-Wale-Elastic-Compression-Stockinet-for-Initial-Stages-of-Refractory-Venous-Leg-Ulcers.pdf
3. Schultz, G. "Wound bed preparation: a systematic approach to wound management." Wound Repair and Regeneration 2003; 11:1–28
4. Sibbald, G. et al. "Special Considerations in Wound Bed Preparation: An Update." Advances in Skin & Wound Care, September 2011
5. Winkler, M. "Does Tissue Response to Fuzzy Wale Elastic Compression Therapy include Cell Micro Deformation, Optimizing Cell Mechano Transduction Signaling to Nuclear DNA to Upregulate Gene Expression & Synthesis of Proteins Required for Healing?" https://edemawear.com/wp-content/uploads/2024/01/PosterDoes-Tissue-Respond-to-Fuzzy-Wale-Elastic-Compression-Therapy092623.pdf
6. Fu, S. et al. "Mechanotransduction in wound healing: from the cellular and molecular level to the clinic." Advances in Skin & Wound Care 34.2 (2021):67-74
7. Saxena, V. "Micro deformations of Wounds and Cell Proliferation," Plastic and Reconstructive Surgery. October 2004, Volume 114, Issue 5,1086-1096

*EdemaWear® Fuzzy Wale Compression™ (FWC) textile, Compression Dynamics LLC, Omaha NE 68102